Preclinical and initial Phase I clinical characterization of CPI-006: an anti-CD73 monoclonal antibody with unique immunostimulatory activity

Piccione EC, Hotston, A, Mikesell G, Daine-Matsuoka B, Gu C, Dao-Pick T, Hill C, Madriaga A, Rudnick J, Liu L, Jones WB, Hammerich L, Brody J, Laport G, Miller RA, Buggy JJ
Corvus Pharmaceuticals, Burlingame, CA 94010, USA.

INTRODUCTION

ADENOSINE IS GENERATED BY CD73 AND CREATES AN IMMUNOSUPPRESSIVE TUMOR MICROENVIRONMENT

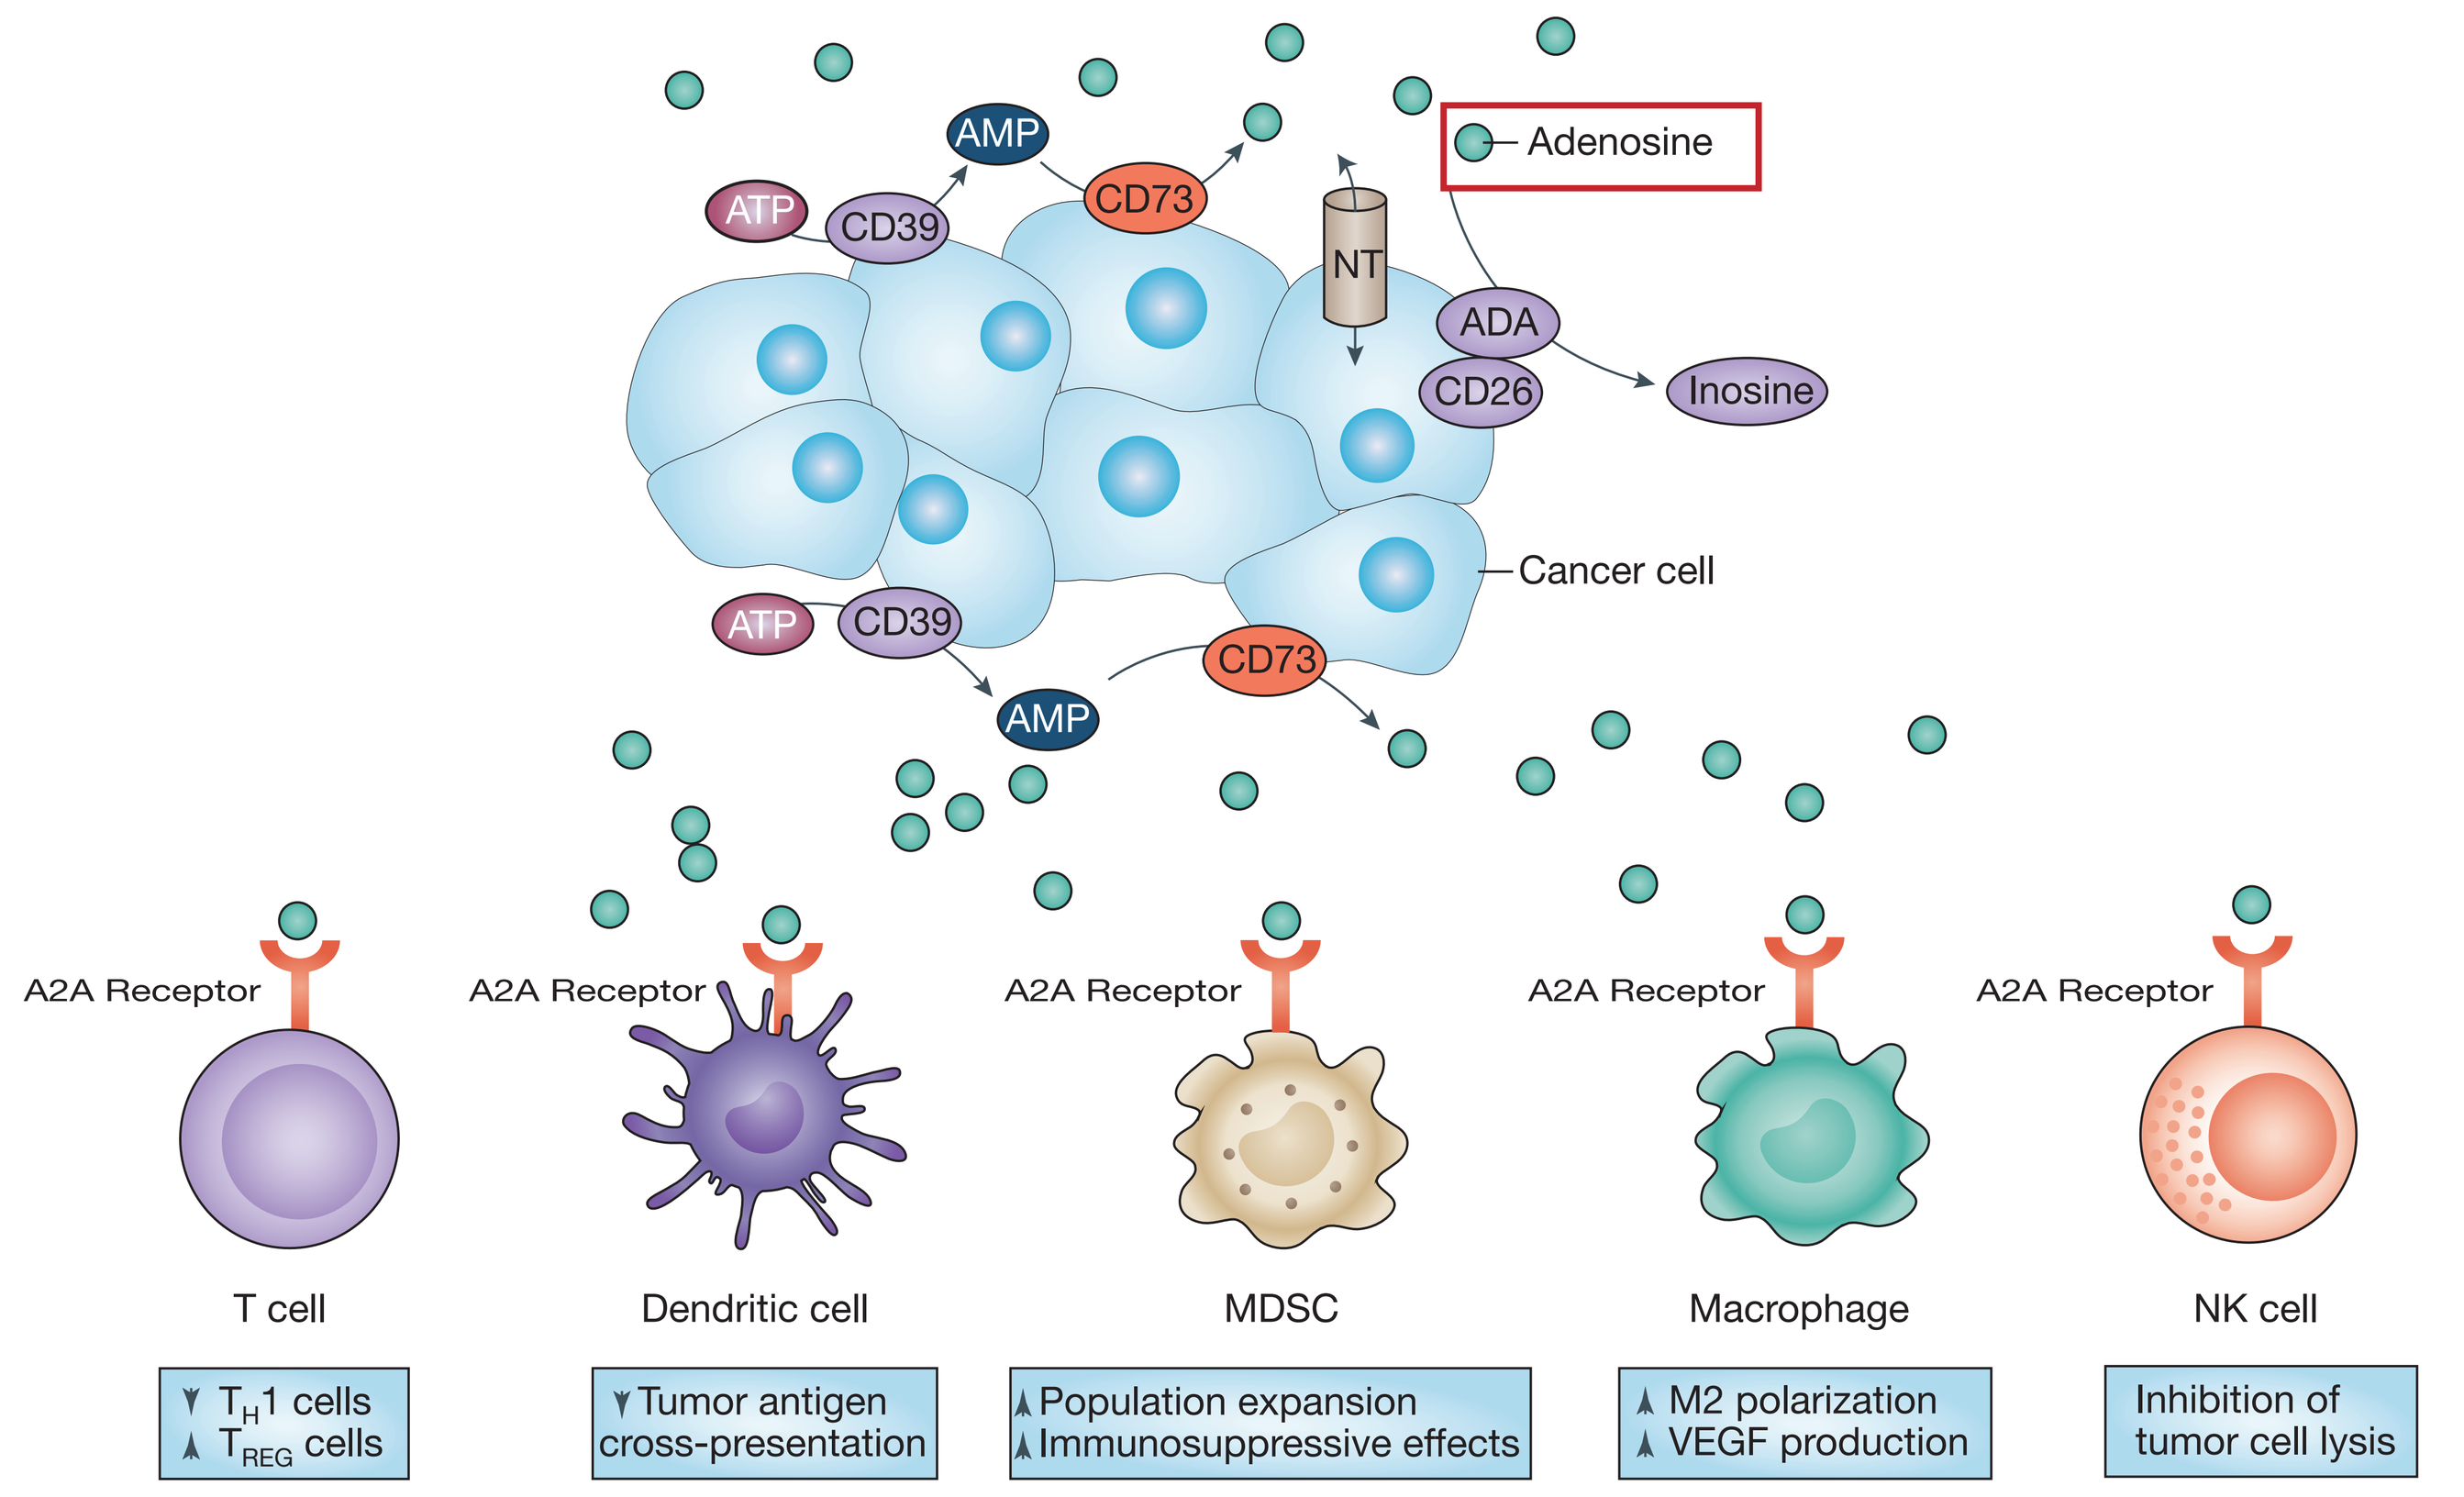
Adenosine
ATP
CD39
AMP
CD73
NT
ADA
CD26
Inosine
Cancer cell
A2A Receptor
T cell
Dendritic cell
MDSC
Macrophage
NK cell
TH1 cells
TREG cells
Tumor antigen cross-presentation
Population expansion
Immunosuppressive effects
M2 polarization
VEGF production
Inhibition of tumor cell lysis

Figure adapted from Antonioli et al, Nat Rev Cancer. 2013

CPI-006 BLOCKS ADENOSINE PRODUCTION AND RELIEVES ADENOSINE-MEDIATED IMMUNOSUPPRESSION

A

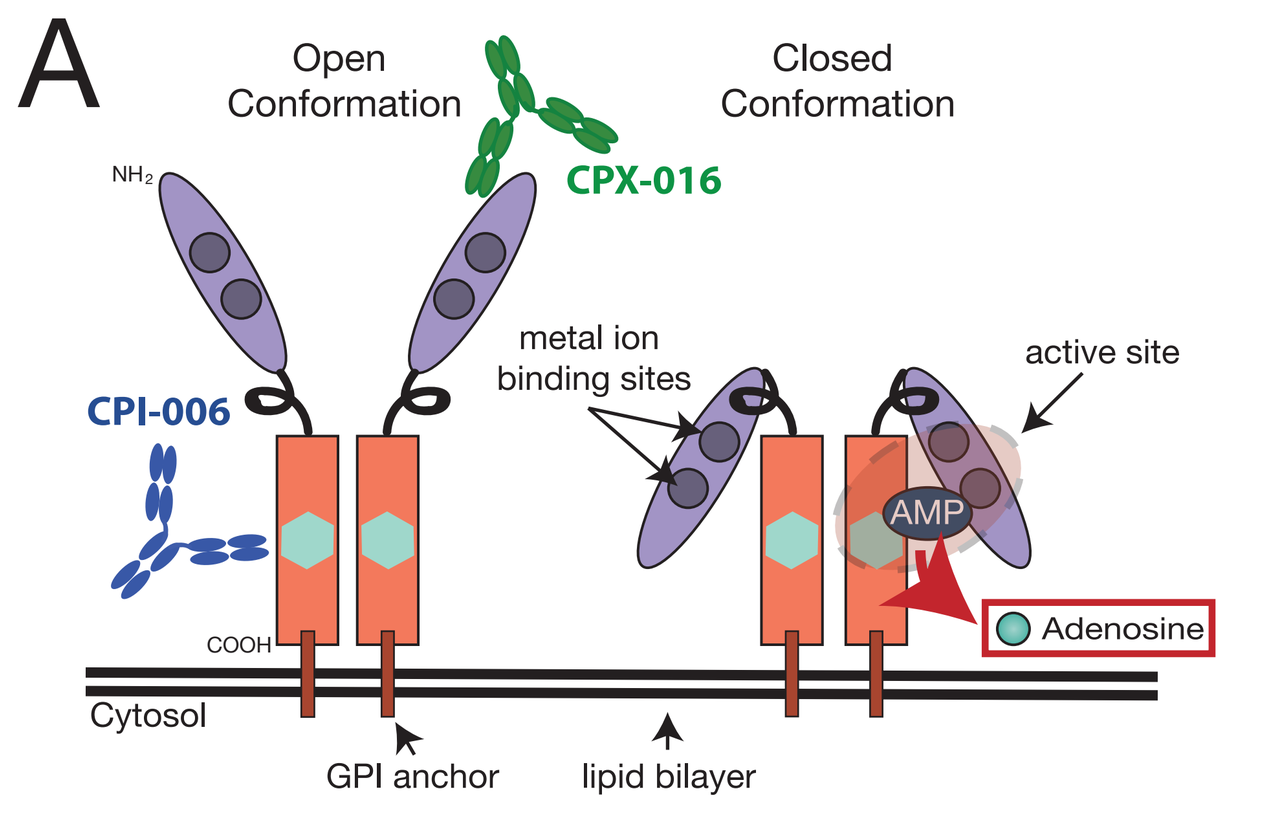
Open Conformation
Closed Conformation
CPX-016
CPI-006
metal ion binding sites
active site
AMP
Adenosine
Cytosol
GPI anchor
lipid bilayer

B CD73 CATALYTIC ACTIVITY

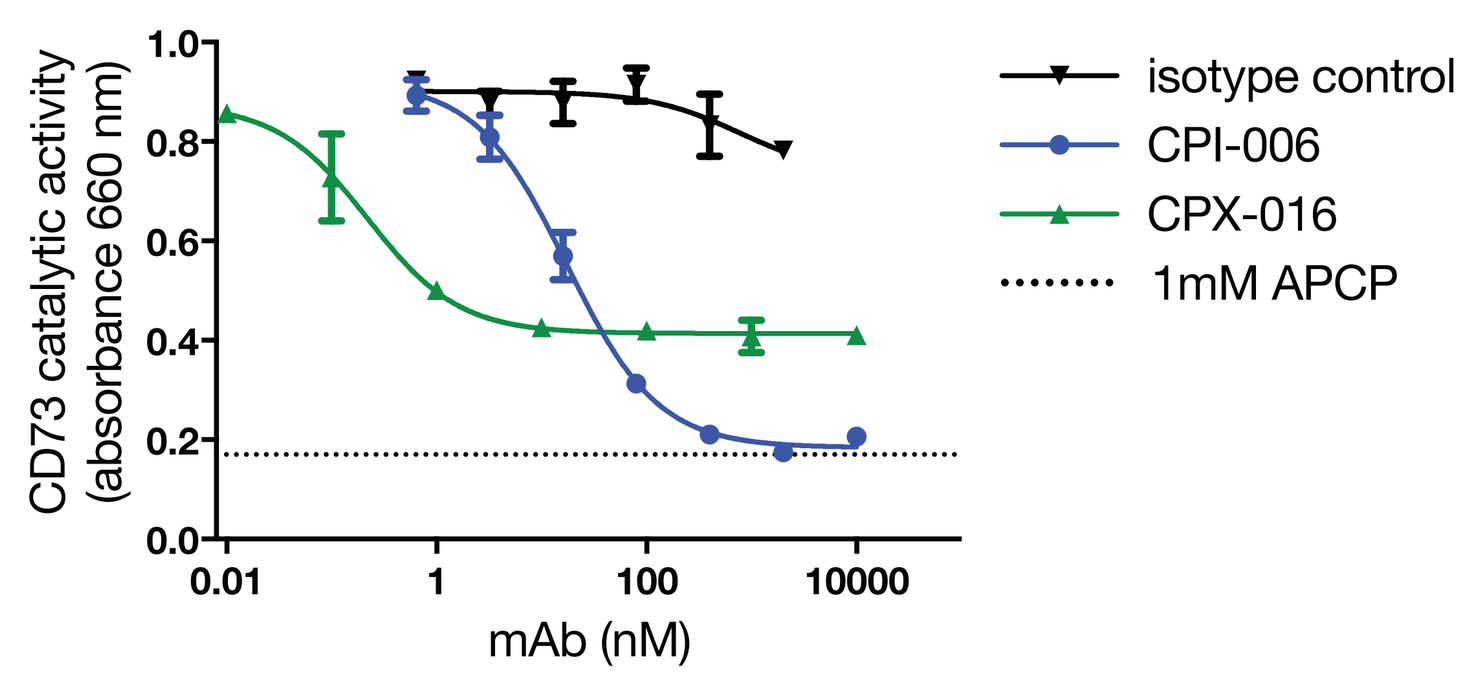
CD73 catalytic activity (absorbance 660 nm)
mAb (nM)
isotype control
CPI-006
CPX-016
1mM APCP

C T CELL PROLIFERATION

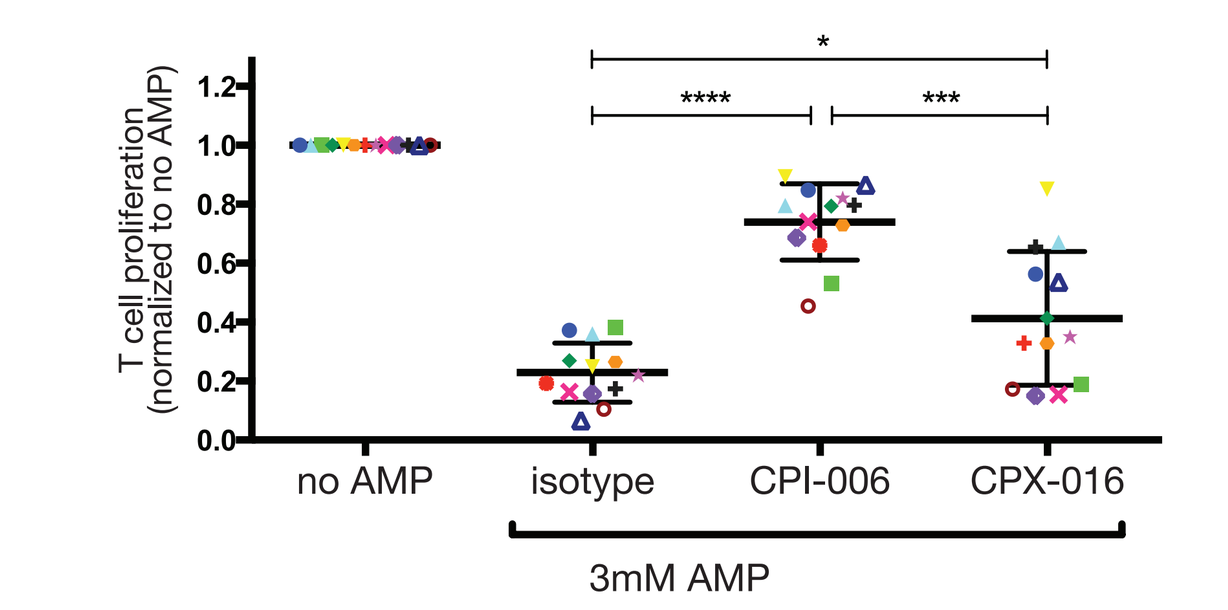
T cell proliferation (normalized to no AMP)
no AMP
isotype
CPI-006
CPX-016
3mM AMP

D IFN-GAMMA SECRETION

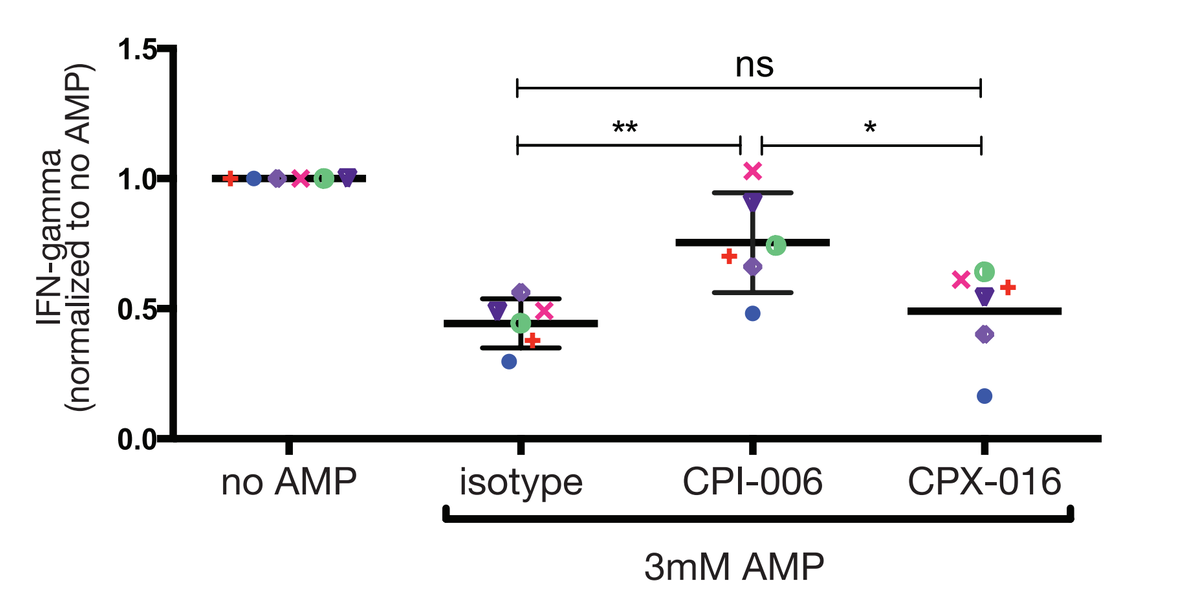
IFN-gamma (normalized to no AMP)
ns
no AMP
isotype
CPI-006
CPX-016
3mM AMP

Figure 1 (A) Schematic of CD73 structure. (B) MDAMB231 cells were incubated with the indicated antibodies or APCP, a small molecule inhibitor of CD73 enzymatic activity, prior to addition of 250 μM AMP. Phosphate levels were measured in the cell culture supernatant using the Sensolyte Malachite Green assay kit. (C,D) PBMC were isolated from healthy donors and labeled with Cell Trace Violet prior to culture with 1 μg/mL anti-CD3 and anti-CD28, 200 units/mL IL-2, 3 mM AMP and indicated treatments (CPI-006, CPX-016 at 500 nM; isotype control at 890 nM). T cell proliferation was measured by Cell Trace Violet dilution and was defined by gating relative to unstimulated PBMC (C). IFN-gamma production was measured in cell culture supernatants by AlphaLISA (D). Each symbol represents an independent donor.

CPI-006 DIRECTLY ACTIVATES HUMAN B LYMPHOCYTES

CPI-006 INDUCES EXPRESSION OF B CELL ACTIVATION MARKERS

A

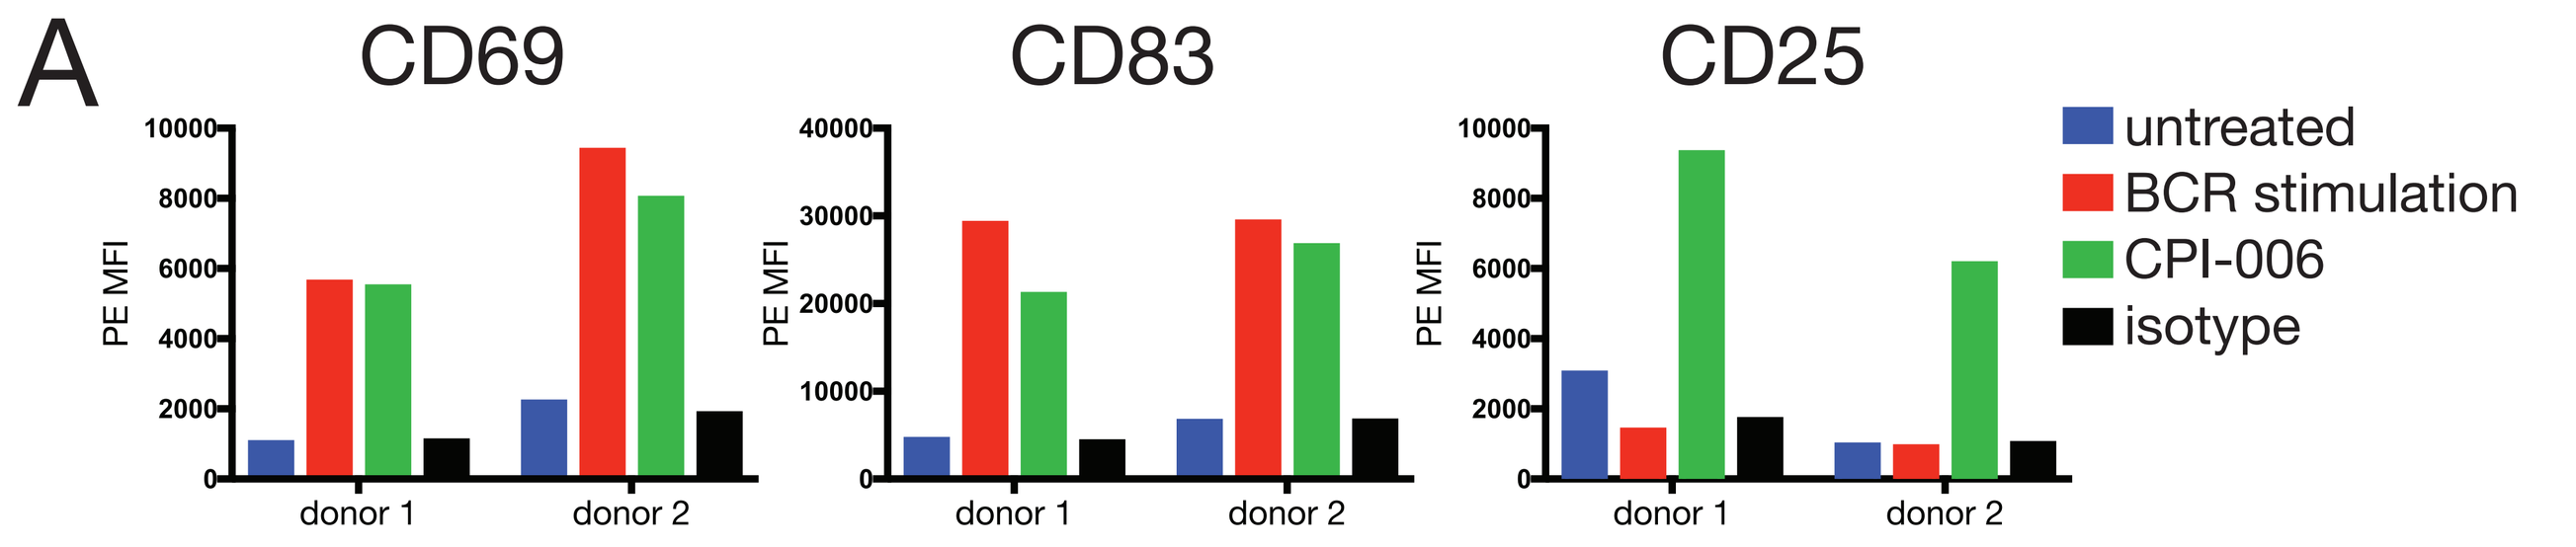
CD69
CD83
CD25
PE MFI
donor 1
donor 2
untreated
BCR stimulation
CPI-006
isotype

INDUCTION OF B CELL ACTIVATION IS UNIQUE TO CPI-006

B

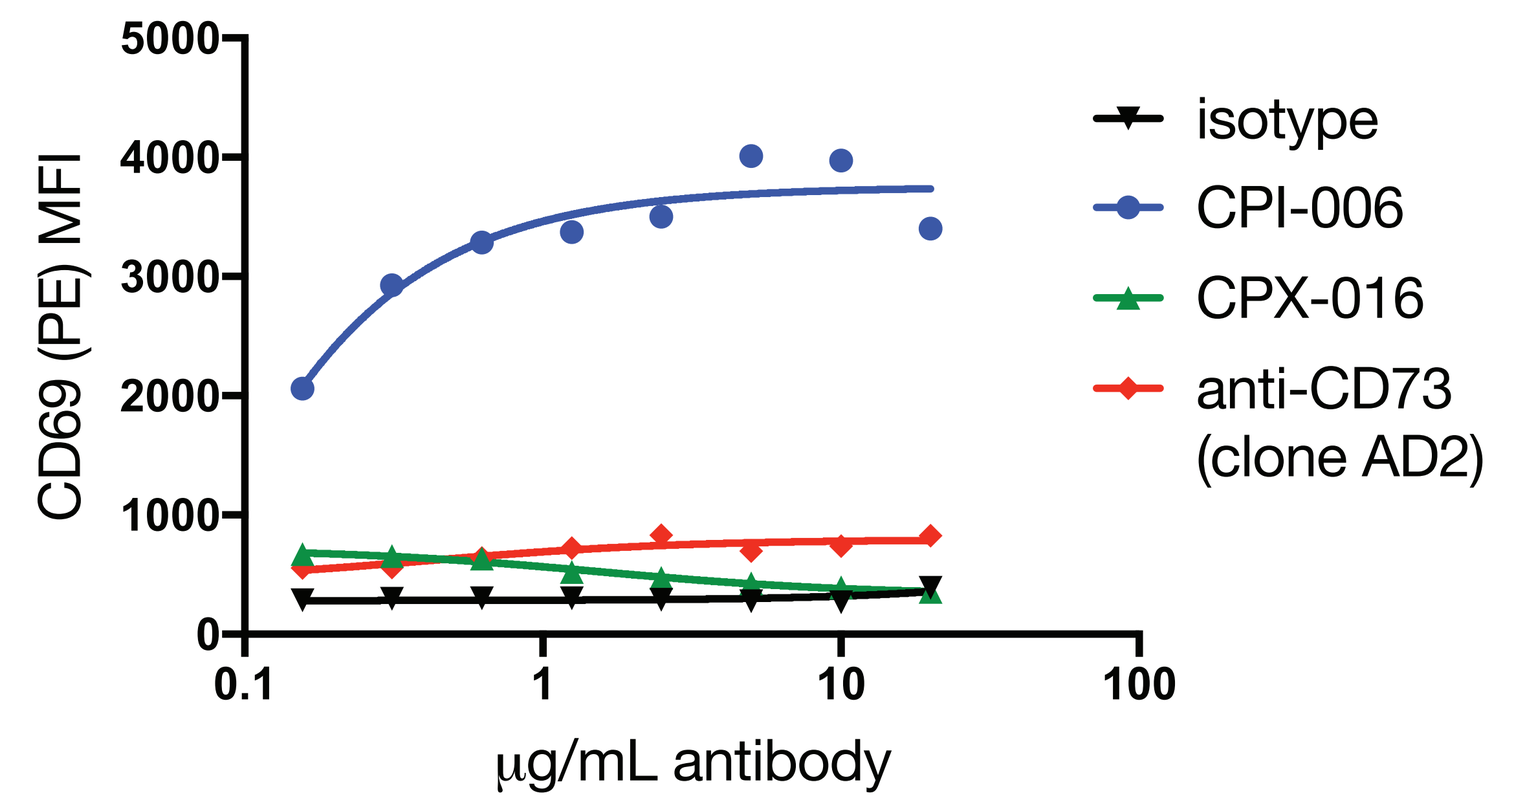
CD69 (PE) MFI
μg/mL antibody
isotype
CPI-006
CPX-016
anti-CD73 (clone AD2)

CPI-006 INDUCES B CELL ACTIVATION VIA BTK

C

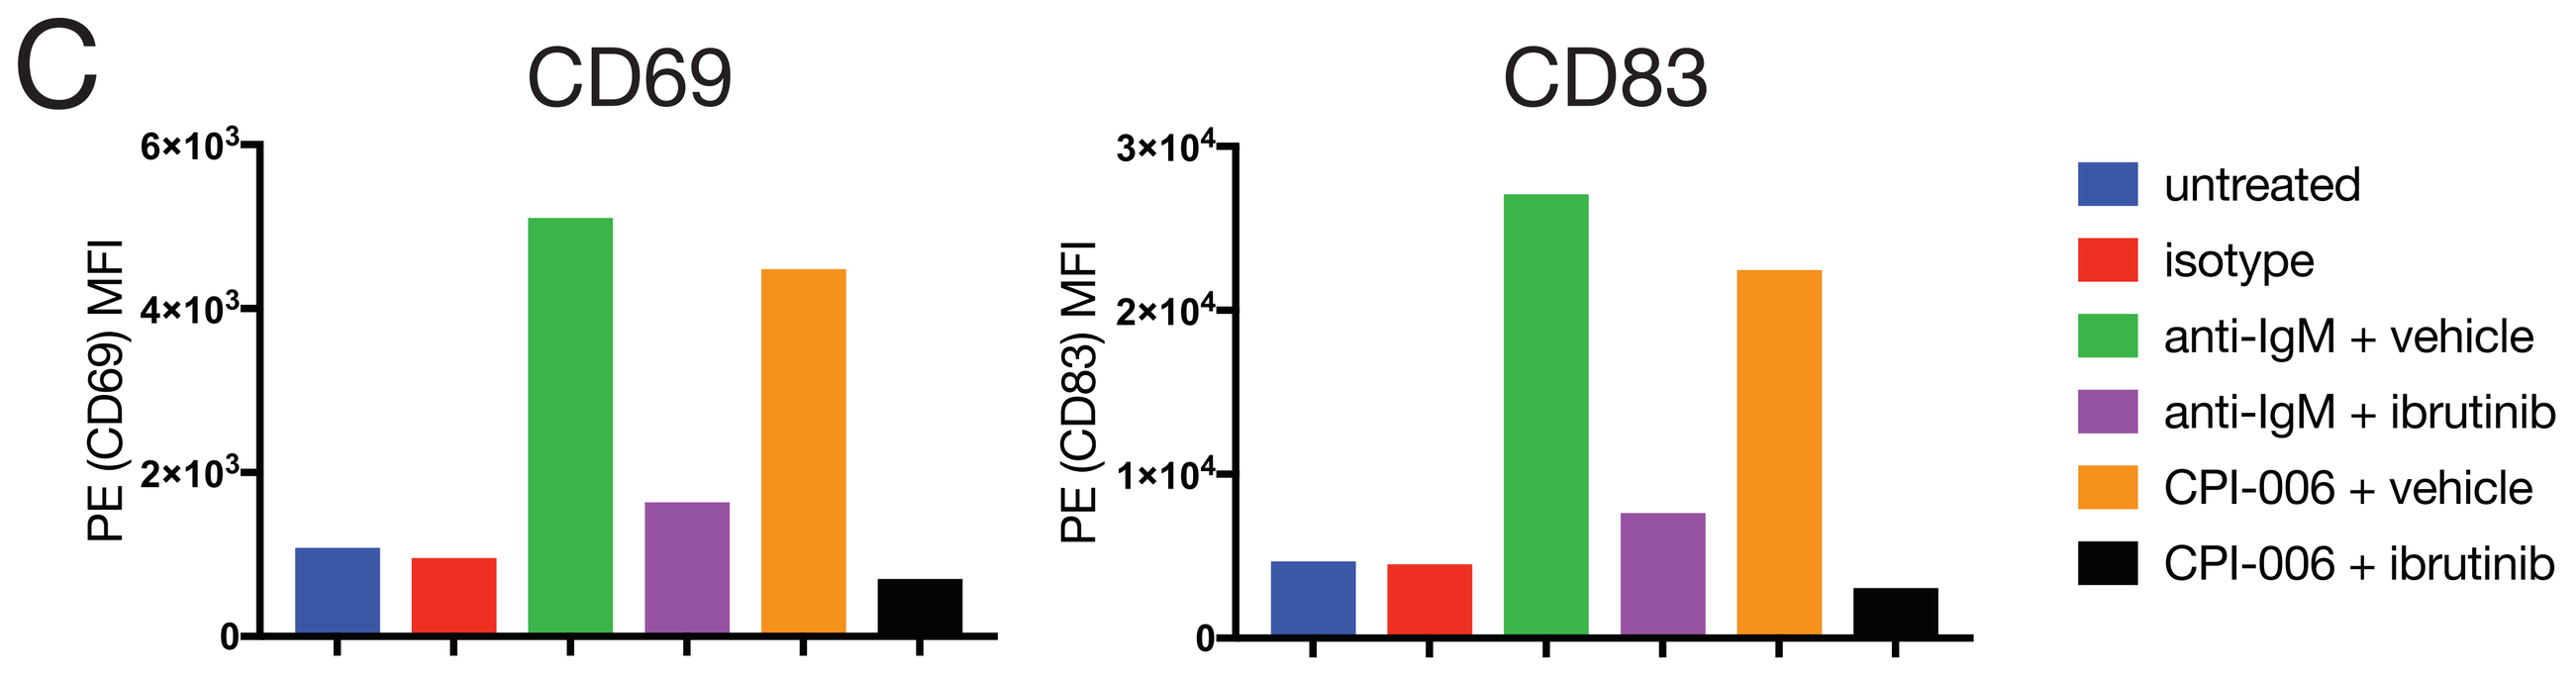
CD69
CD83
PE (CD69) MFI
PE (CD83) MFI
untreated
isotype
anti-IgM + vehicle
anti-IgM + ibrutinib
CPI-006 + vehicle
CPI-006 + ibrutinib

Figure 2 (A) PBMC were isolated from healthy donors and incubated with B cell receptor (BCR) stimulation (bead-bound anti-IgM), CPI-006 (10 μg/mL), or human IgG1 isotype control (10 μg/mL) overnight. Flow cytometry analysis was performed with gating on B cells (CD19+CD3-) and mean fluorescence intensity (MFI) is reported for staining with antibodies to the indicated cell surface markers. (B) PBMC were isolated from healthy donors and incubated with the indicated treatments over a range of concentrations. Flow cytometry analysis was performed with gating on B cells (CD19+CD3-) and mean fluorescence intensity (MFI) is reported for antibody staining of CD69. (C) PBMC were isolated from healthy donors and incubated overnight with bead-bound anti-IgM or CPI-006 (1 μg/mL) +/- BTK inhibitor, ibrutinib (100 nM). Flow cytometry analysis was performed with gating on B cells (CD19+CD3-) and mean fluorescence intensity (MFI) is reported for antibody staining of CD69 and CD83.

CPI-006 DIRECTLY ACTIVATES HUMAN B LYMPHOCYTES

CPI-006 INDUCES B CELL ACTIVATION INDEPENDENT OF ADENOSINE

D

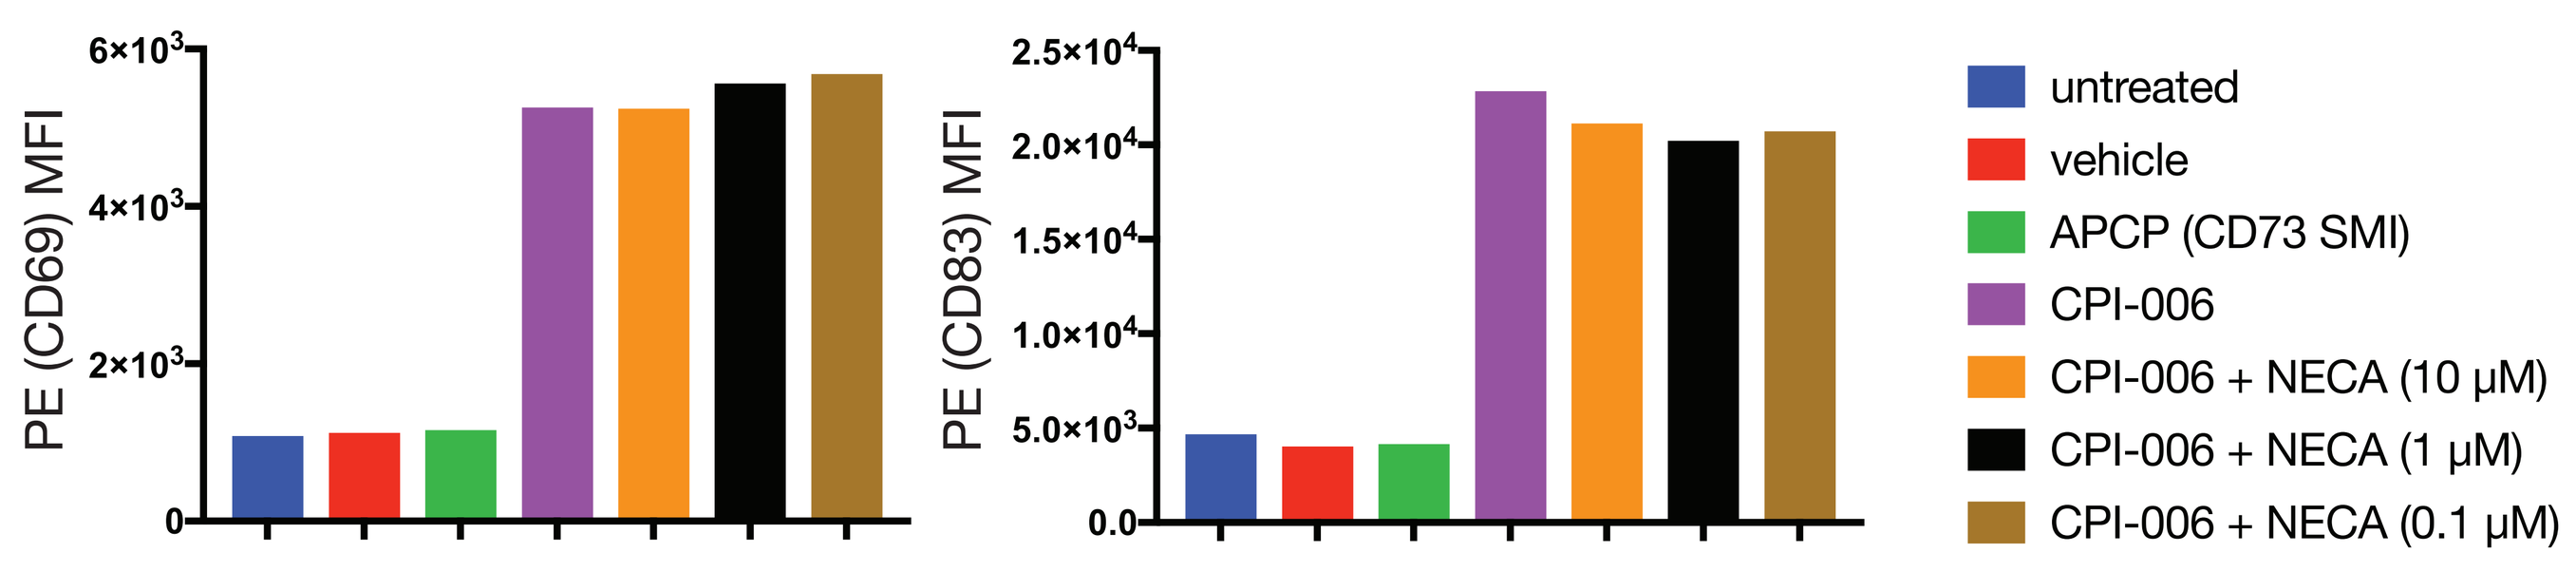
CD69
CD83
PE (CD69) MFI
PE (CD83) MFI
untreated
vehicle
APCP (CD73 SMI)
CPI-006
CPI-006 + NECA (10 μM)
CPI-006 + NECA (1 μM)
CPI-006 + NECA (0.1 μM)

CPI-006 INDUCES PHOSPHO-ERK SIGNALING IN B CELLS

E

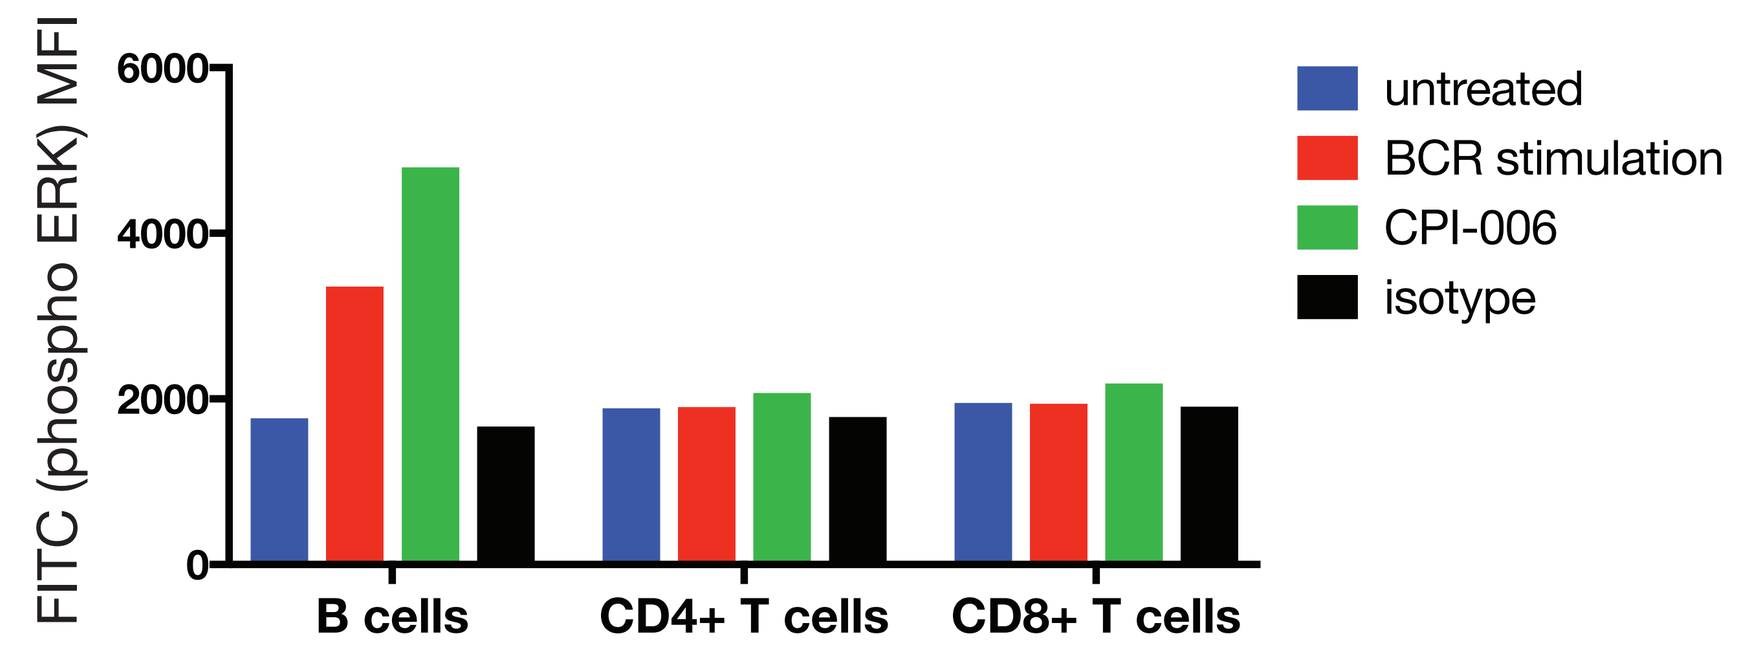
FITC (phospho ERK) MFI
B cells
CD4+ T cells
CD8+ T cells
untreated
BCR stimulation
CPI-006
isotype

Figure 2, continued (D) PBMC were isolated from healthy donors and incubated overnight with APCP (1μM) or CPI-006 (1 μg/mL) +/- NECA. Flow cytometry analysis was performed with gating on B cells (CD19+CD3-) and mean fluorescence intensity (MFI) is reported for antibody staining of CD69 or CD83. (E) PBMC were isolated from healthy donors and incubated with the indicated treatments for 15 minutes. Flow cytometry analysis was performed with gating on the indicated cell populations and staining of phosho ERK was evaluated.

PHASE 1/1B CLINICAL TRIAL DESIGN

DOSE ESCALATION
3+3 DESIGN: 1, 3, 6, 12, 18, 24 MG/KG

CPI-006 alone | CPI-006 + CPI-444 (A2AR antagonist) | CPI-006 + pembrolizumab

DOSE EXPANSION
STAGE 1 (N=11 PER COHORT)

CPI-006 alone | CPI-006 + CPI-444 (A2AR antagonist) | CPI-006 + pembrolizumab

NSCLC | RCC | Others — NSCLC | RCC | Others — NSCLC | RCC | Others

If 1 or more responses observed in a disease cohort, proceed to Stage 2

DOSE EXPANSION
STAGE 2 (N=17 PER COHORT)

PHARMACOKINETICS AND PHARMACODYNAMICS AFTER FIRST INFUSION

SERUM PHARMACOKINETICS AND RECEPTOR OCCUPANCY

A

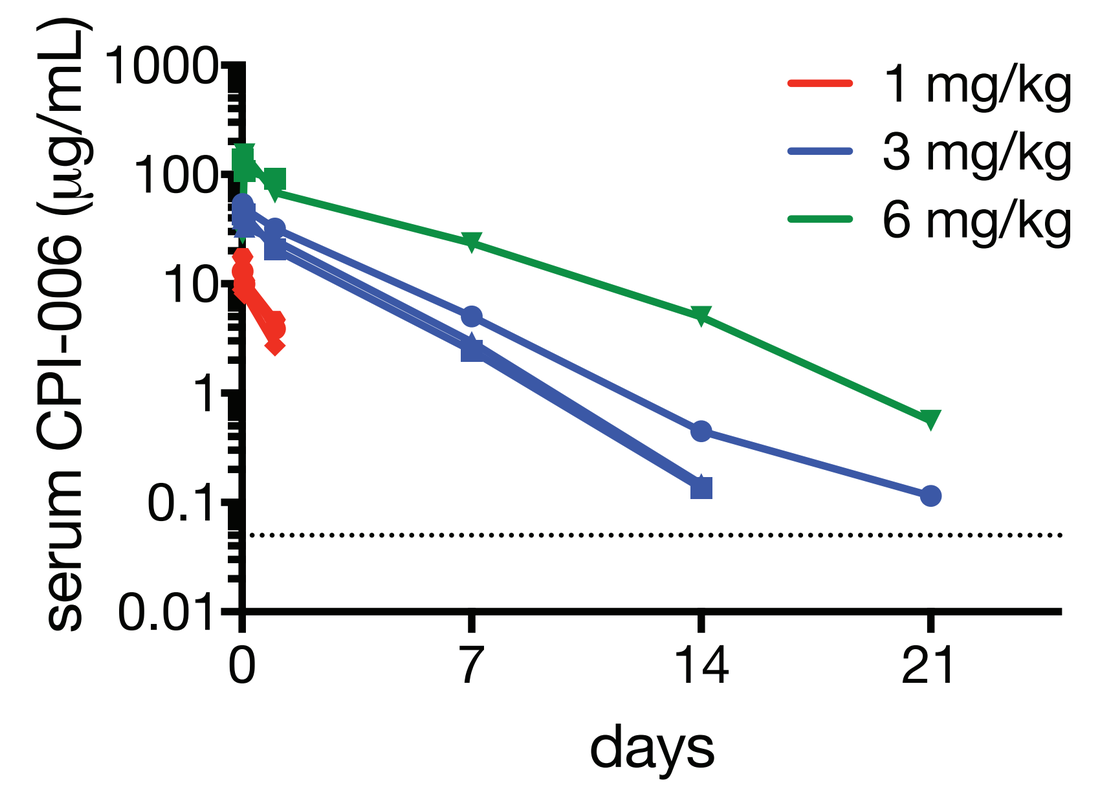
serum CPI-006 (μg/mL)
days
1 mg/kg
3 mg/kg
6 mg/kg

B

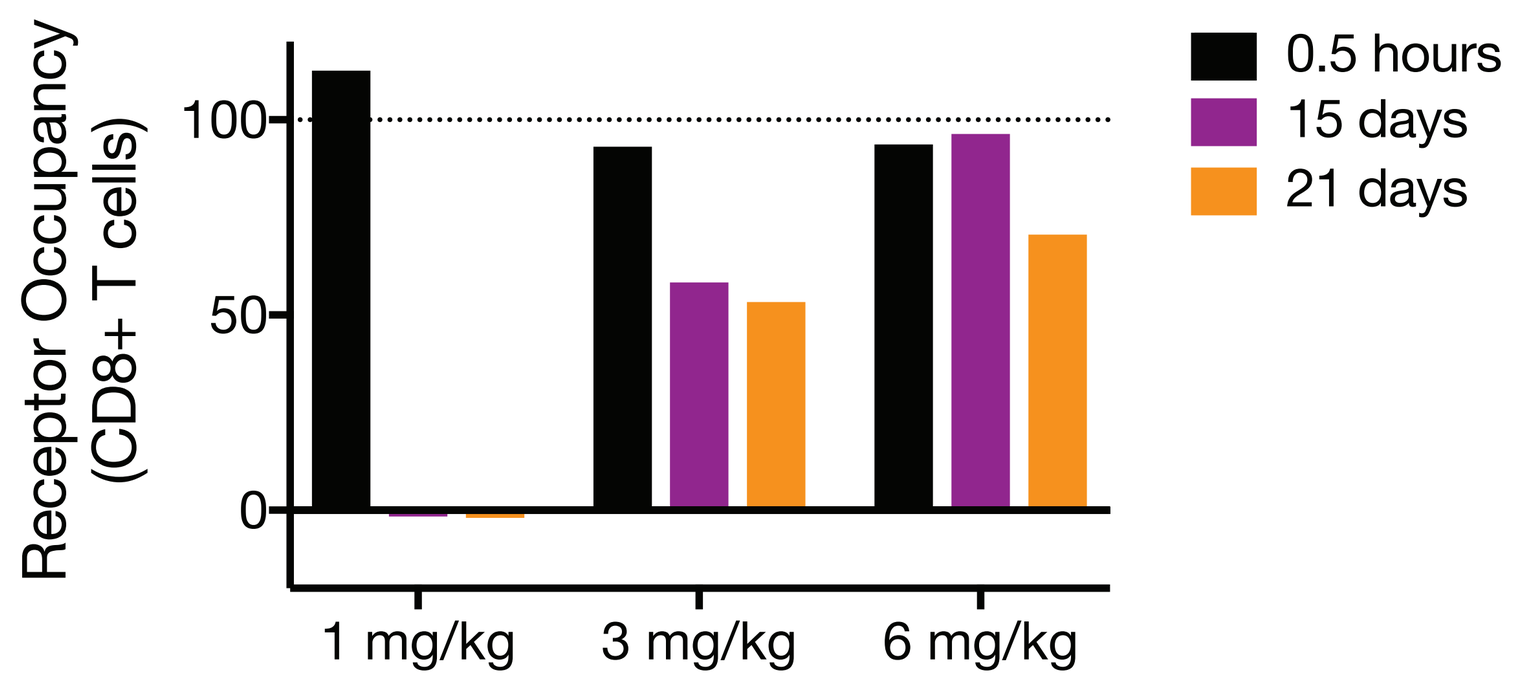
Receptor Occupancy (CD8+ T cells)
1 mg/kg
3 mg/kg
6 mg/kg
0.5 hours
15 days
21 days

Patient	Dose	Disease	Occupancy of Peripheral B cells at C1D15	% Decrease of Peripheral B cells at C1D1 (0.5h)	Serum CPI-006 [μg/mL] at C1D8
1	1 mg/kg	bladder	-1.35%	24.6%	BLQ
2		prostate	65.83%	82.6%	BLQ
3		SCHN	22.96%	63.7%	BLQ
4	3 mg/kg	pancreatic	88.12%	71.2%	2.461
5		pancreatic	74.57%	62.3%	5.032
6		prostate	58.19%	68.1%	2.937
7	6 mg/kg	colorectal	97.90%	49.3%	23.60
8		prostate	98.31%	64.6%	TBD
9		SCHN	101.60%	11.1%	TBD

No Grade 3/4 Adverse Events were observed

Figure 3 (A) Serum samples were collected from patients treated with CPI-006 monotherapy and levels of free CPI-006 were measured by ELISA. (B) Whole blood samples from patients treated with CPI-006 monotherapy were fixed and receptor occupancy was measured by flow cytometry gating on CD73+ CD8 T cells.

CPI-006 TRANSIENTLY REDISTRIBUTES PERIPHERAL B CELLS

A

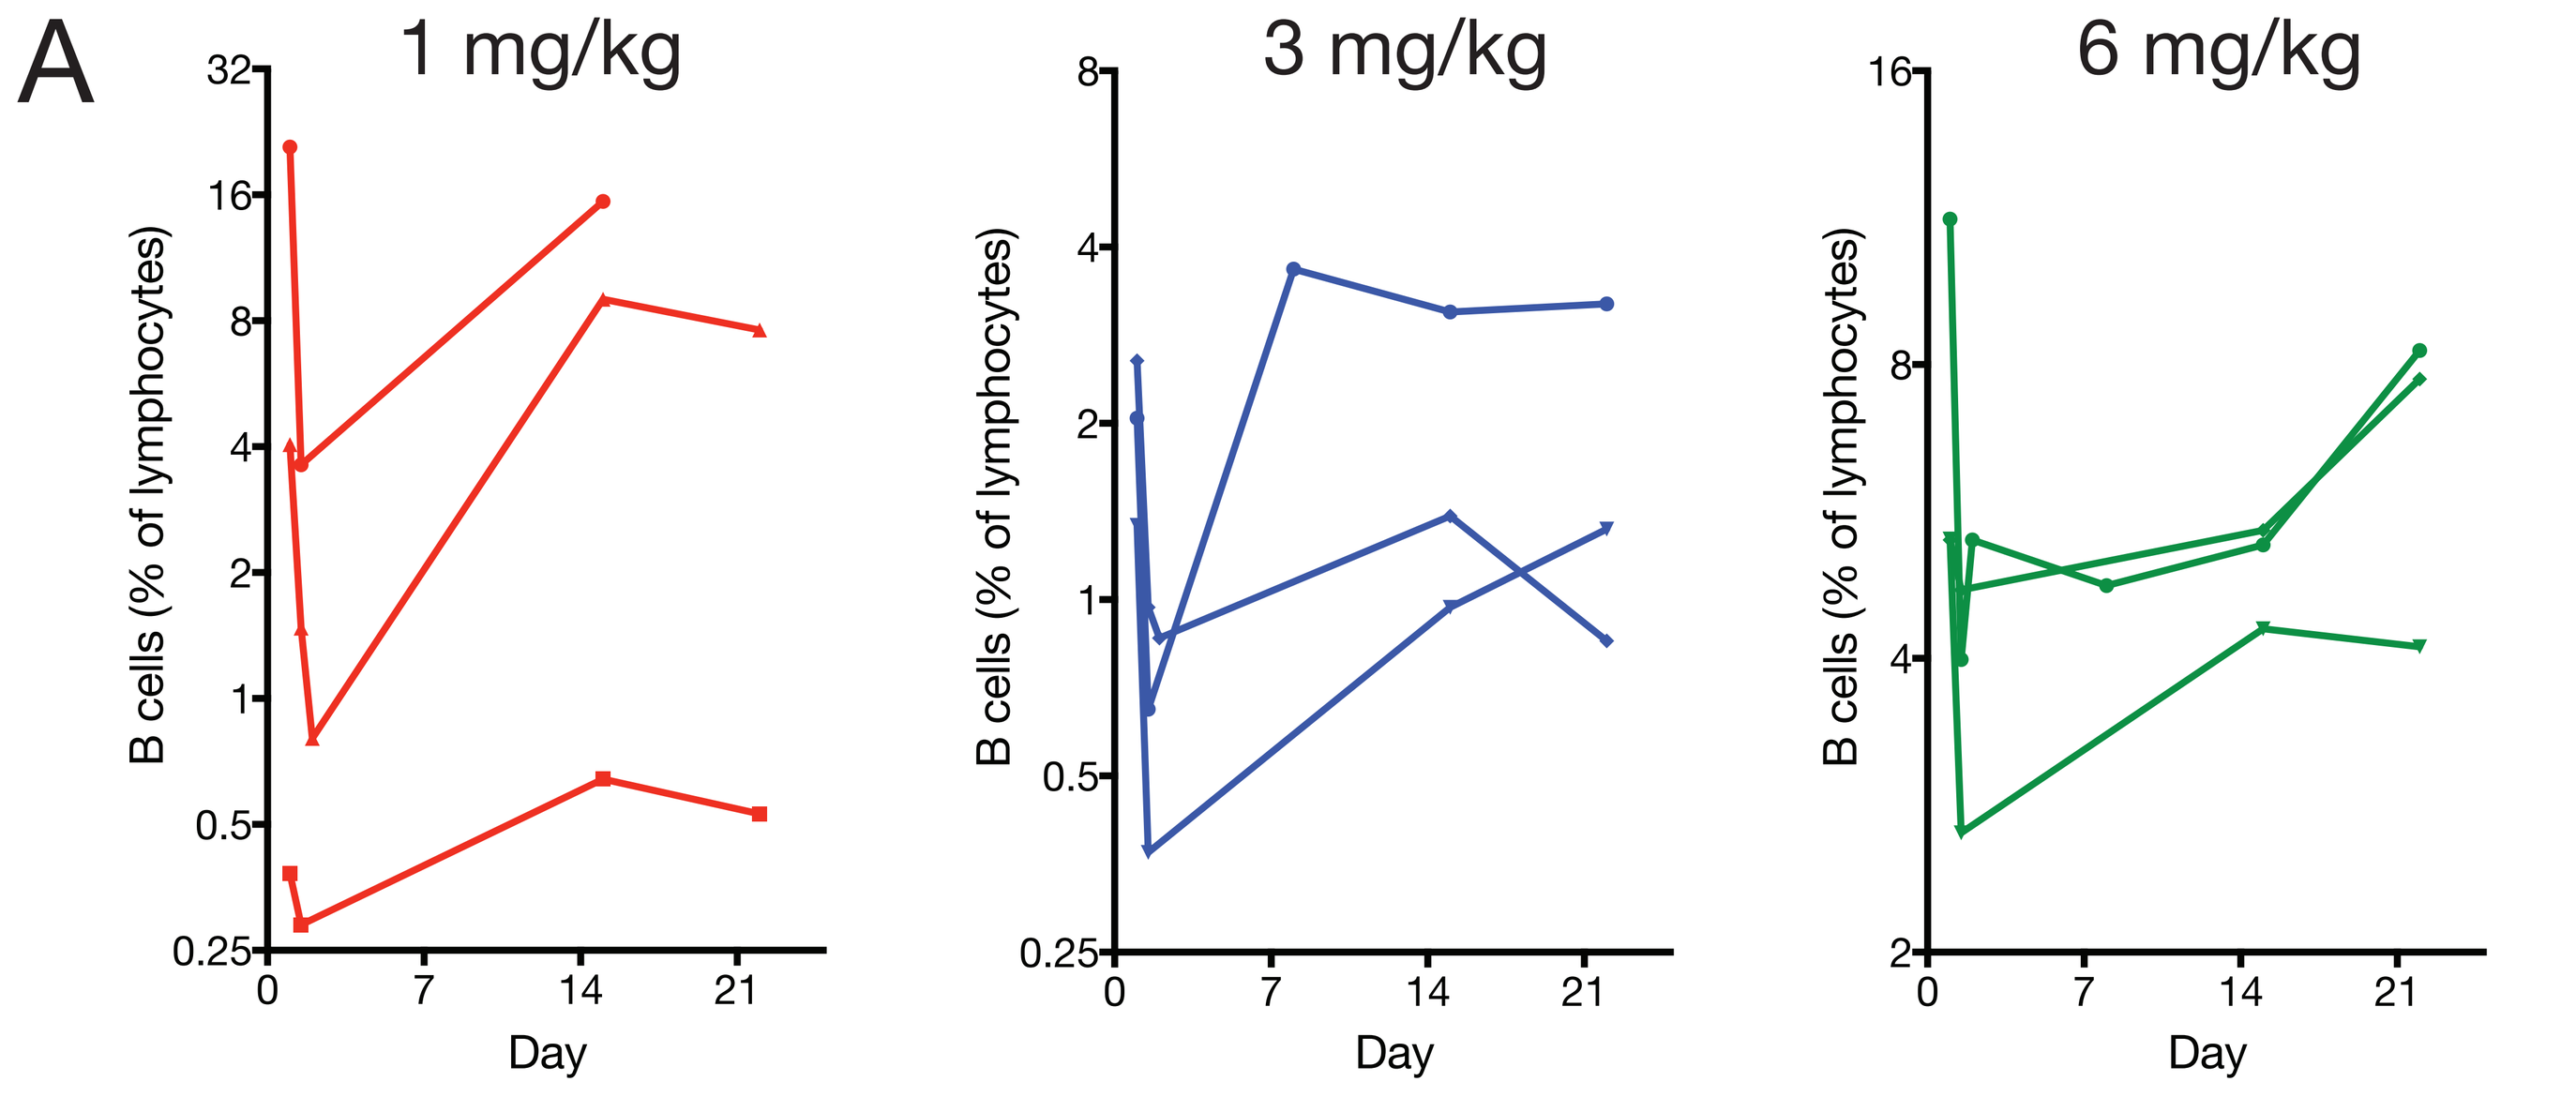
1 mg/kg
3 mg/kg
6 mg/kg
B cells (% of lymphocytes)
Day

B

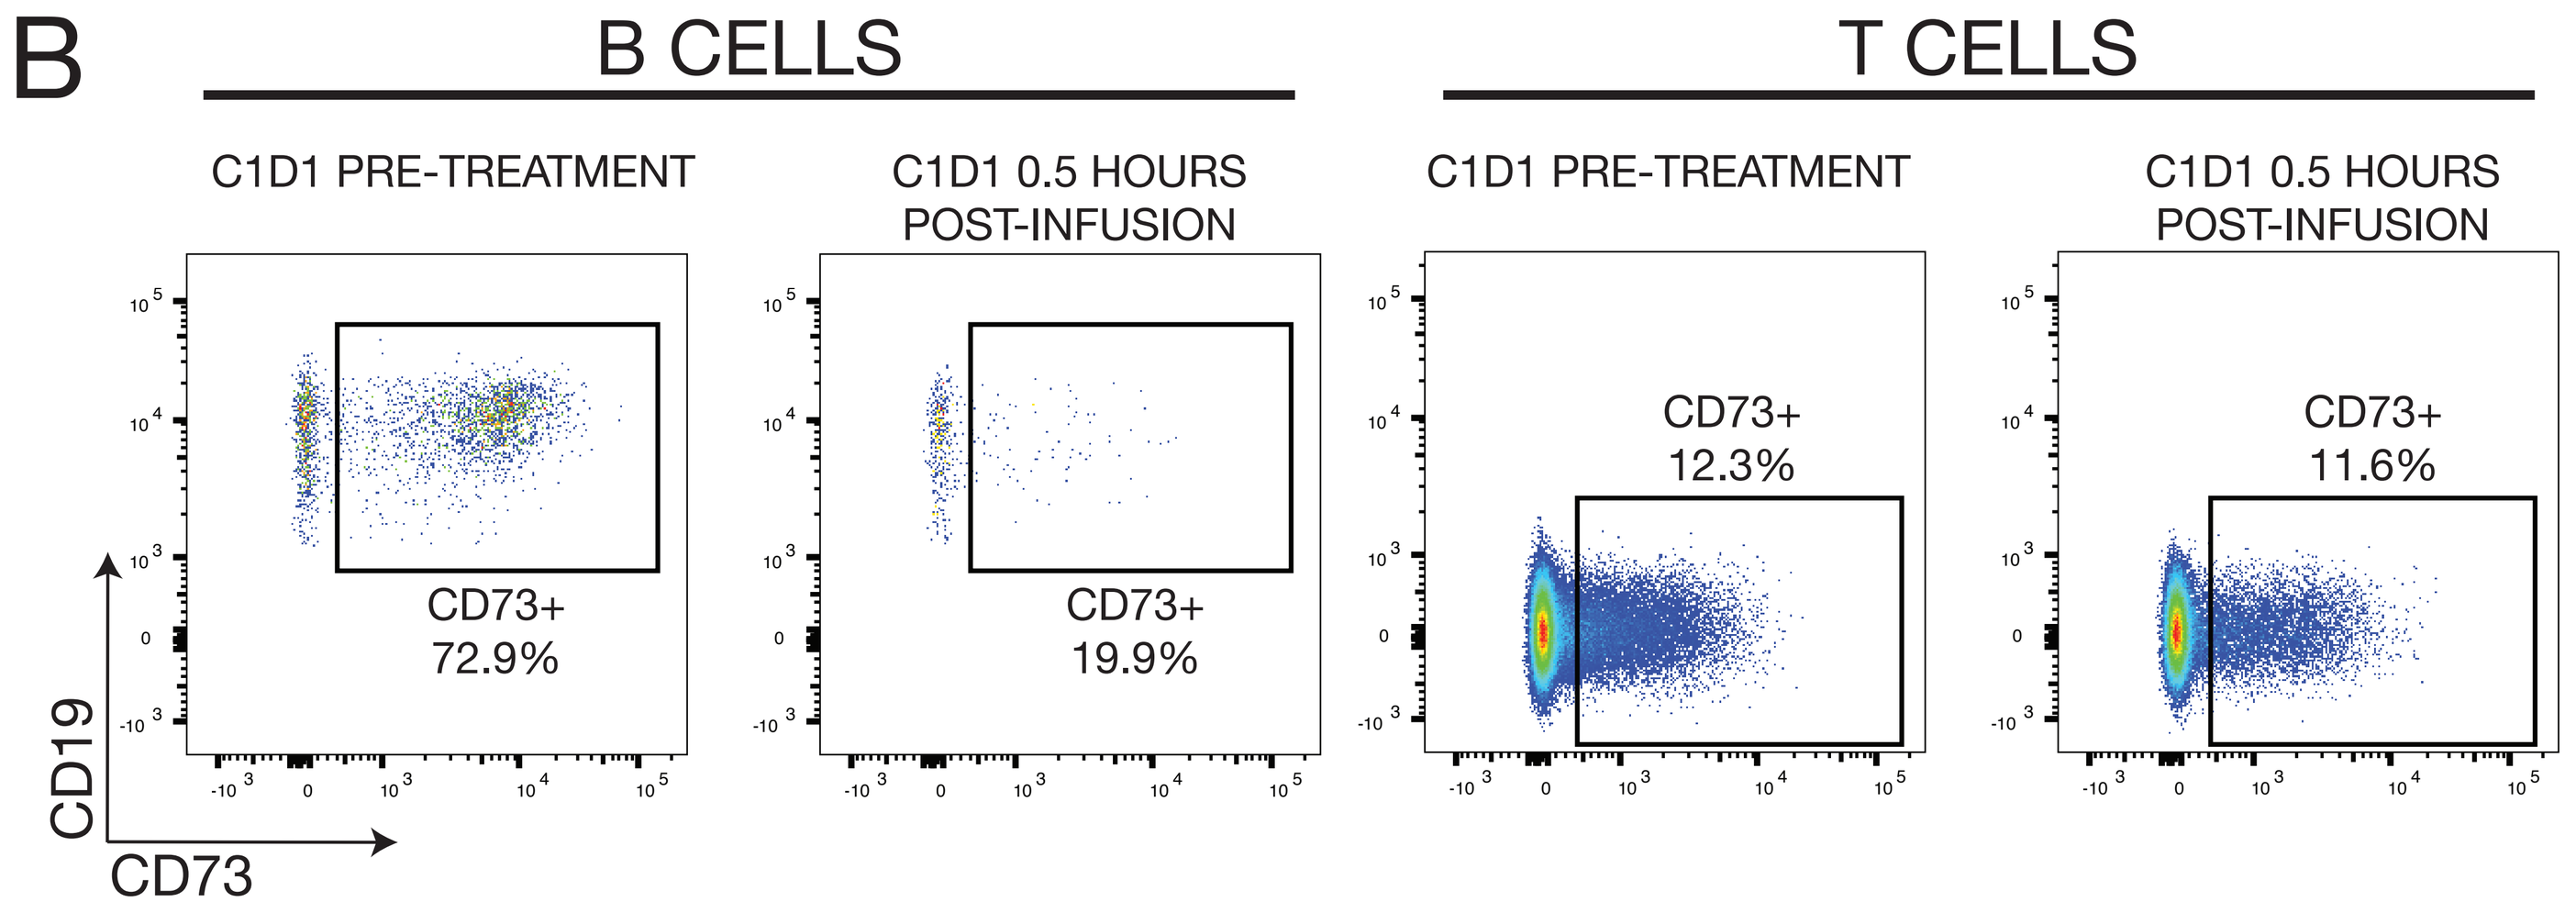
B CELLS
T CELLS
C1D1 PRE-TREATMENT
C1D1 0.5 HOURS POST-INFUSION
CD73+ 72.9%
CD73+ 19.9%
CD73+ 12.3%
CD73+ 11.6%
CD19
CD73

C

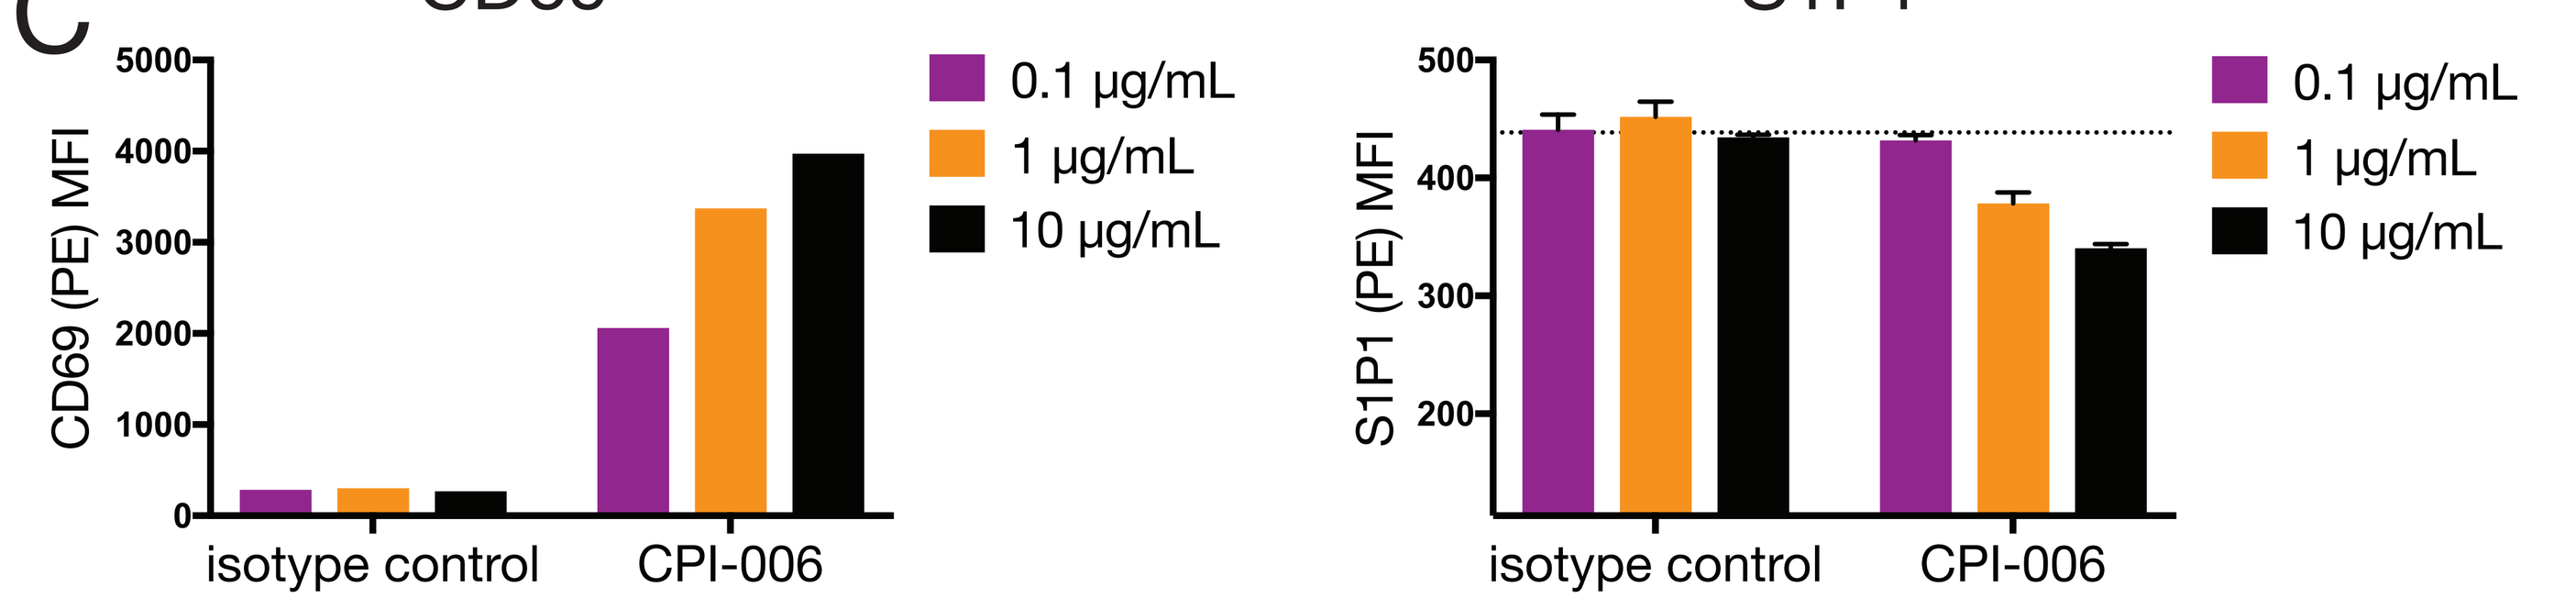
CD69
S1P1
CD69 (PE) MFI
S1P1 (PE) MFI
0.1 μg/mL
1 μg/mL
10 μg/mL
isotype control
CPI-006

D

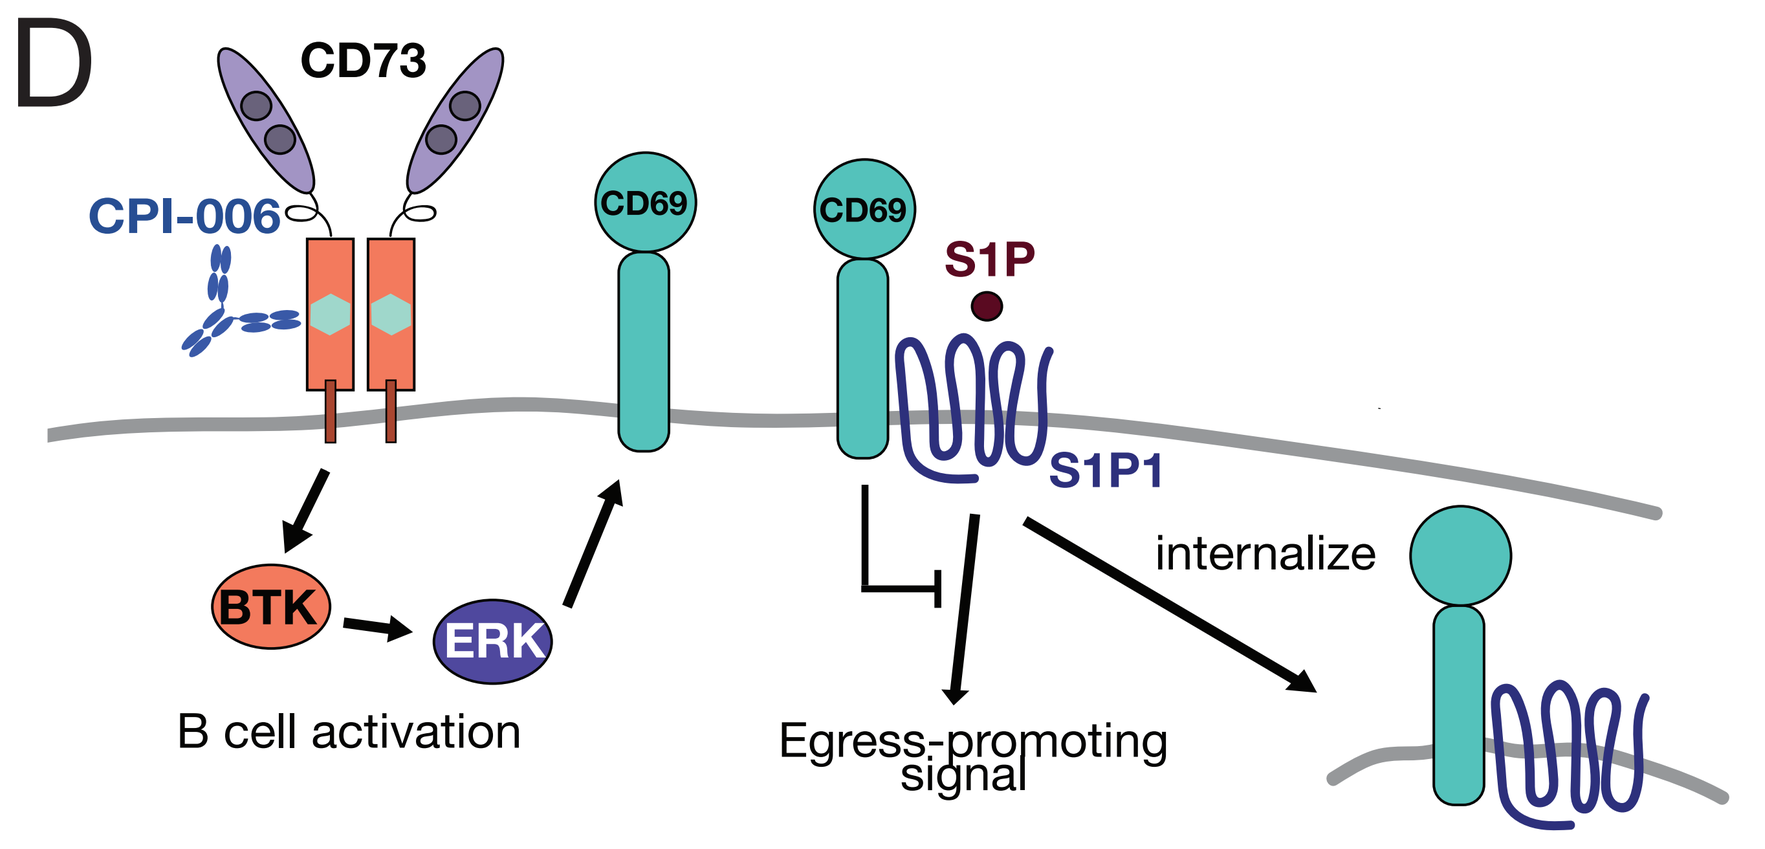
CD73
CPI-006
CD69
S1P
S1P1
BTK
ERK
B cell activation
Egress-promoting signal
internalize

- S1P is abundant in the blood and peripheral lymphocytes have low surface levels of its receptor, S1P1
- S1P1 is uregulated on lymphocytes after migration into lymphoid organs
- Surface expression of S1P1 is required for signaling that promotes egress
- Downmodulation of S1P1 blocks egress from lymphoid organs

Figure 4 (A,B) Whole blood samples from patients treated with a single dose of CPI-006 monotherapy were evaluated by flow cytometry. (A) Levels of B cells (CD19+CD3-) are reported as a percent of total lymphocytes (gated based on scatter properties). (B) CD73 expression was evaluated with a non-competing anti-CD73 antibody and is reported on B cells (CD19+CD3-) and T cells (CD19-CD3+). (C) Purified B cells from healthy human donors were incubated with CPI-006 or isotype control at the indicated concentrations for 30 minutes. Surface levels of CD69 and S1P1 were determined by flow cytometry with gating on CD73+ B cells. (D) A model for the mechanism leading to reduction in levels of peripheral B cells by CPI-006. CPI-006 binding to CD73+ B cells induces expression of CD69, which promotes internalization of S1P1. S1P1 signaling mediates egress of lymphocytes from lymphoid tissues and S1P1 internalization promotes retention in lymphoid organs. [Shiow et al, Nature (2006) Vol 440, 540-544] [Lo et al, JEM (2005) Vol 201, 291-301]

CONCLUSIONS

- CPI-006 targets a novel epitope on CD73
 - Blocks production of adenosine by inhibiting the enzymatic active site
 - Activates B cells, leading to increased expression of CD69
- Phase I monotherapy clinical data indicates that CPI-006 is:
 - Well tolerated at doses evaluated so far: 1, 3, 6 mg/kg with no DLTs
 - Dose proportional PK and receptor occupancy observed
 - Affects B lymphocyte trafficking as shown by transient redistribution of B cells
- Dose escalation continues with monotherapy and combination with A2AR antagonist CPI-444